Characterization of reversible dysfunctions in regulatory CD8 T cell population responsible for pathogenic T cell elimination in T1D patients

MOZART THERAPEUTICS
Orchestrating The Immune System

Daniel T. Patton, Jennifer L. Gardell, Meghan E. Maurer, Tara N. McCray, Nadine N. Morgan, Monica M. Childs, Susan H. Julien, Cong Tan, Catherine J. McMahan, Courtney A. Crane, Kristine M. Swiderek

BACKGROUND/AIMS:

- Regulatory CD8 T cells (CD8 Treg) have been described as regulators of immune homeostasis and are characterized by oligoclonal T cell receptors (TCRs) directed toward conserved peptide sequences; they appear dysfunctional in patients with autoimmune diseases
- CD8 Treg reduce disease in adoptive transfer studies and are increased in patients with improved outcomes in response to immunotherapies, including in teplizumab and alefacept treated type 1 diabetes (T1D) patients
- CD8 Treg function by selectively killing pathogenic T cells that promote tissue destruction, without broad immunosuppression
- This study sought to define the phenotype and functional characteristics of CD8 Treg isolated from patients with T1D

METHODS

- CD8 Treg from the peripheral blood of T1D patients and from healthy donors were comparatively assessed using flow cytometry
- Functional responses of T1D patient-derived CD8 Treg were evaluated using pan TCR stimulation (anti-CD3) or T1D autoantigen peptide cocktail
- Effects on pathogenic T cells were evaluated using peptide restimulation of peripheral blood mononuclear cells (PBMC)
- Effects on tissue damage were tested using functional human pancreatic islet-derived organoids in the presence of T1D patient PBMC

Model of Dysfunctional CD8 Treg Network

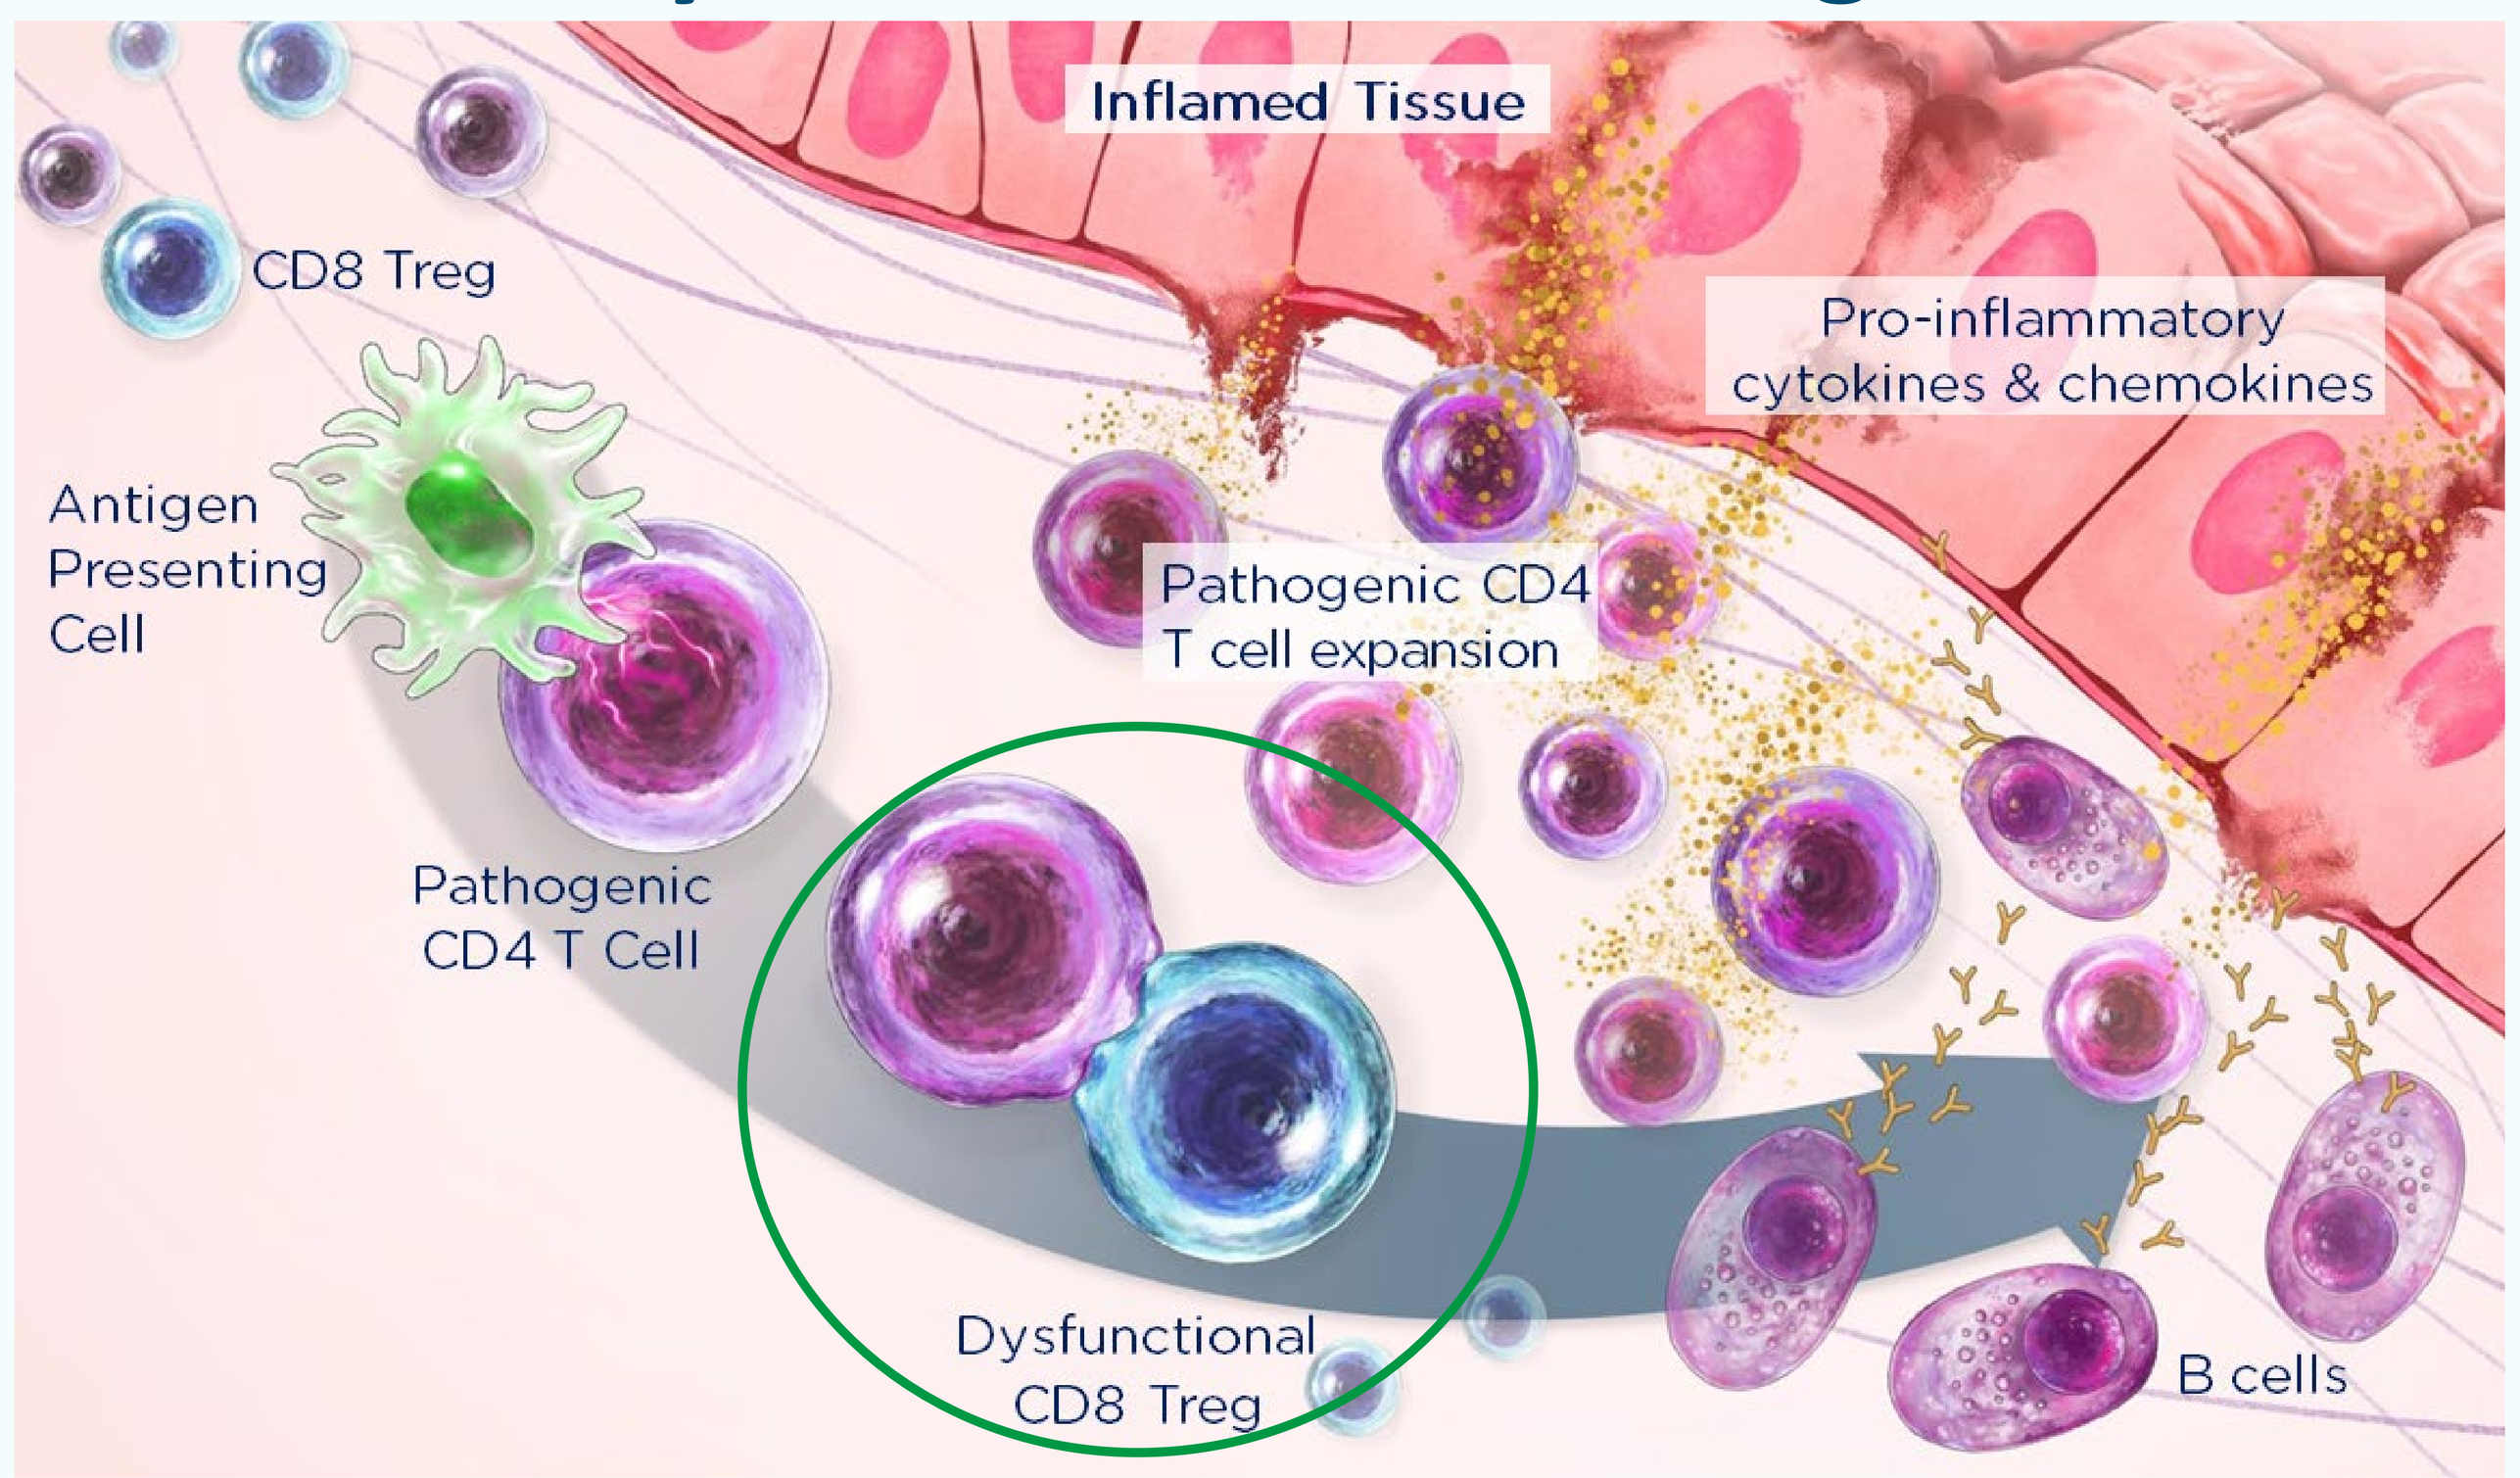
Inflamed Tissue
CD8 Treg
Pro-inflammatory cytokines & chemokines
Antigen Presenting Cell
Pathogenic CD4 T cell expansion
Pathogenic CD4 T Cell
Dysfunctional CD8 Treg
B cells

Figure 1. Model of a dysfunctional CD8 Treg network. When CD8 Treg fail to eliminate pathogenic CD4 T cells, CD4 T cells can expand, secrete proinflammatory cytokines, recruit and activate other inflammatory cells (e.g. pathogenic CD8 T cells), and induce autoantibody production. Collectively, this cascade results in the destruction of healthy tissue.

Dysfunctional CD8 Treg are Reduced Yet Present and Highly Responsive to TCR Stimulation in Patients More Than 5 Years After Diagnosis of Type 1 Diabetes

(A) CD8 Treg prevalence and activation in T1D PBMCs

(B) Responses to TCR activation

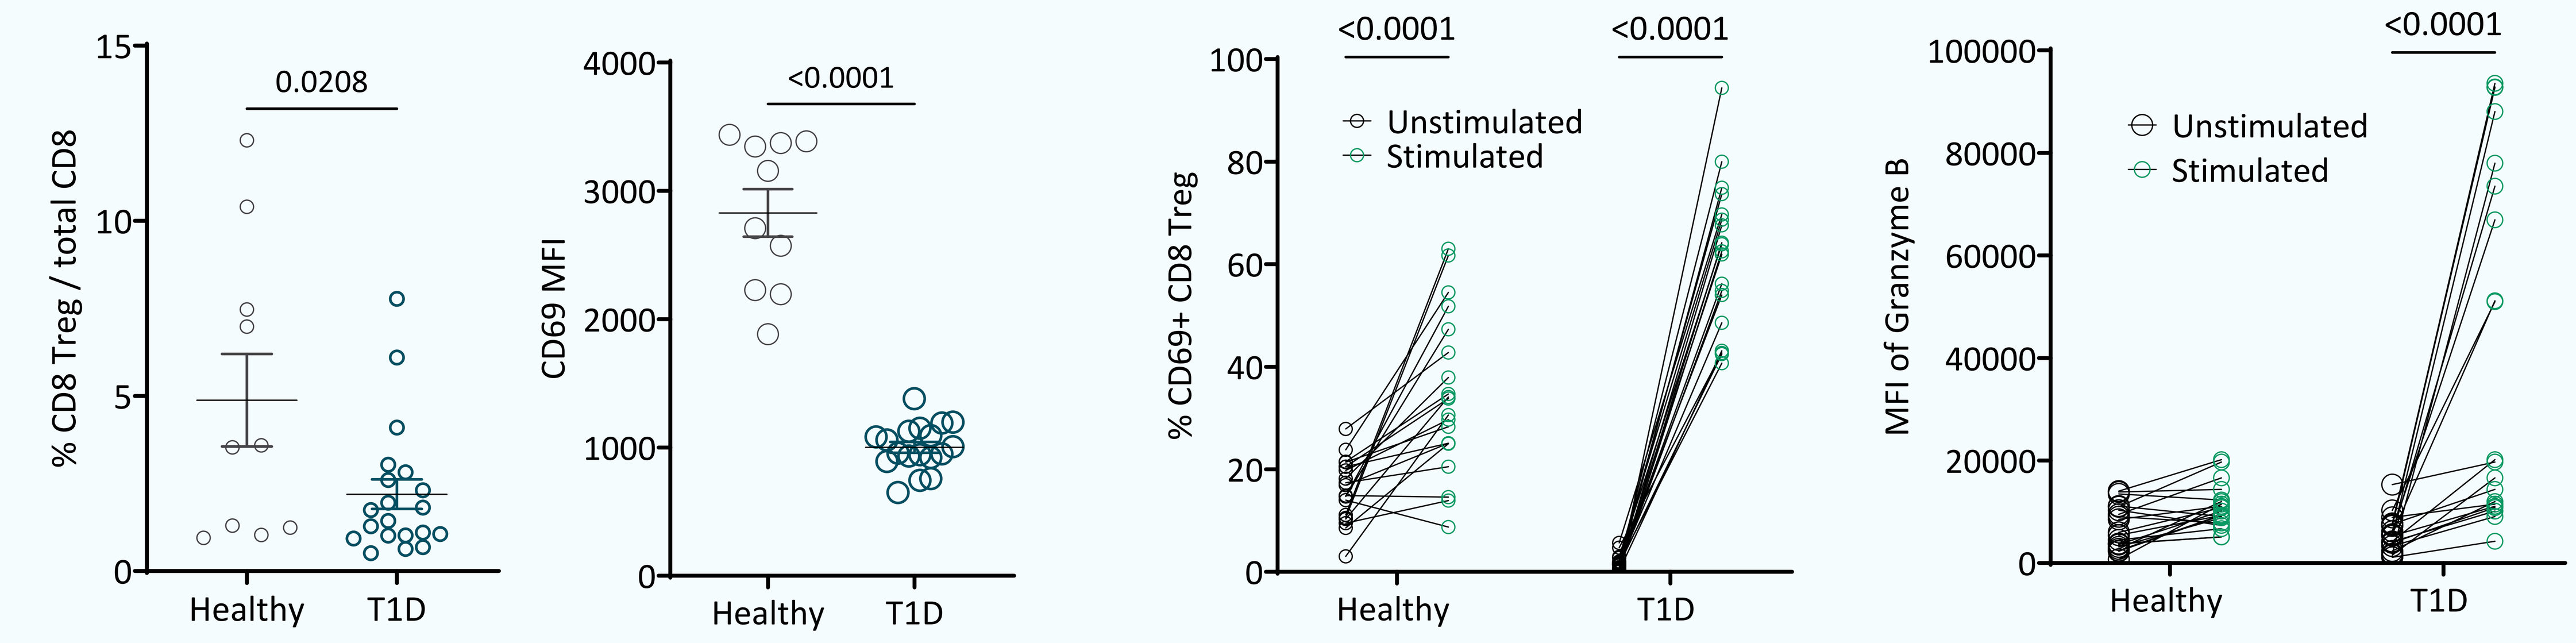
% CD8 Treg / total CD8
0.0208
Healthy
T1D
CD69 MFI
<0.0001
% CD69+ CD8 Treg
Unstimulated
Stimulated
MFI of Granzyme B

Figure 2: CD8 Treg in T1D patient PBMC collected more than 5 years since diagnosis are dysfunctional, but remain highly responsive to TCR stimulation (A) Proportion of CD8 Treg out of the total CD8 T cell population, and their activation status using flow cytometry analysis of prevalence and activation status (CD69). (B) PBMC from healthy (n=10) or T1D donors (n=20) were cultured for 24 hours in anti-CD3 and activation status and cytolytic capacity (Granzyme B) were assessed.

Antigenic Peptides Induce CD4 T Cell Activation in T1D PBMC

(A) CD4 T cell activation **(B) IFNγ ELISPOT**

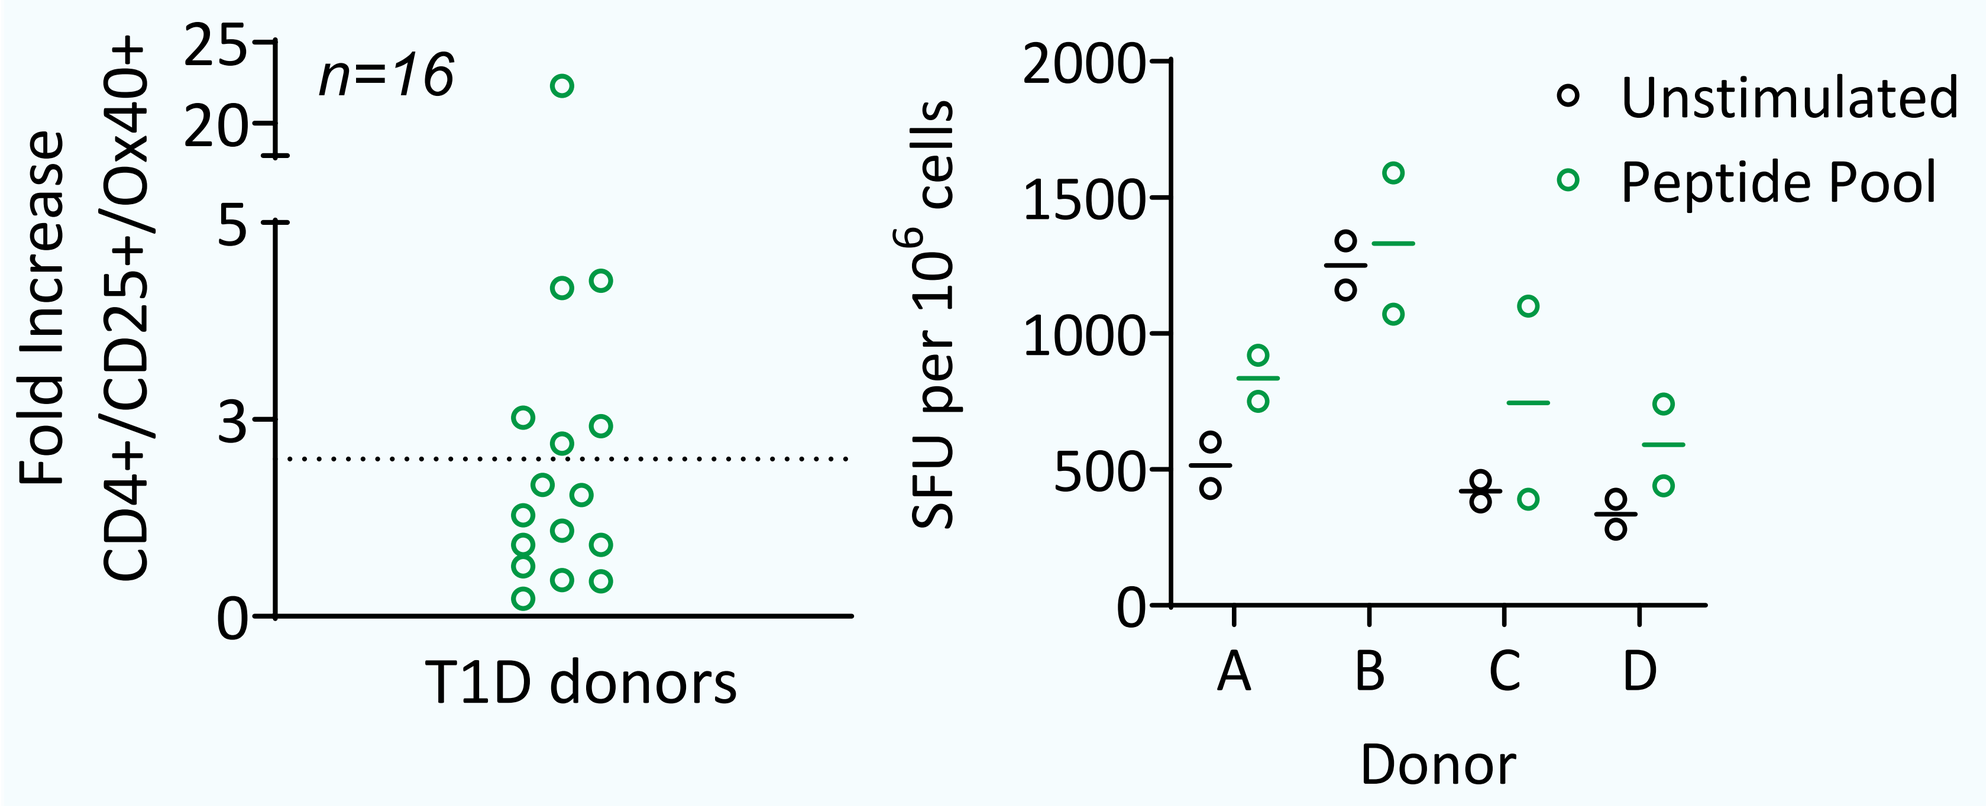
Fold Increase CD4+/CD25+/Ox40+
n=16
T1D donors
SFU per 10^6 cells
Unstimulated
Peptide Pool
A B C D
Donor

Figure 3: Long term diagnosed T1D patients retain autoantigen recall responses. T1D patient PBMC were stimulated with a pool of 11 peptides derived from GAD65, IA2 and PPI. (A) Increase in proportion of $CD4^+CD25^+Ox40^+$ cells relative to unstimulated. PBMC showing >2x fold increase were judged to be responders. (B) Number of IFNγ spot forming units (SFU) in 4 of the responding patients from (A).

Human Pancreatic Organoids Contain Relevant Cell Populations with Expected Localization

(A) Establishment of Pancreatic Organoids

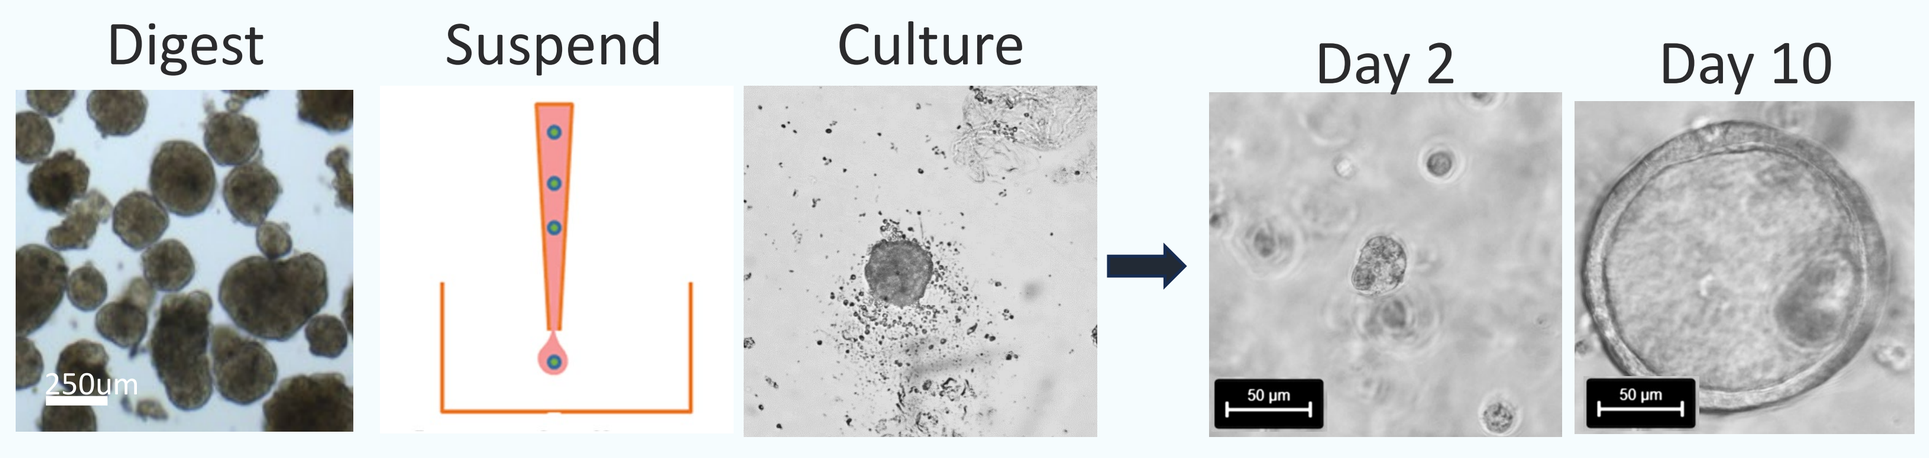
Digest
Suspend
Culture
Day 2
Day 10

(B) Characterization of Protein Expression

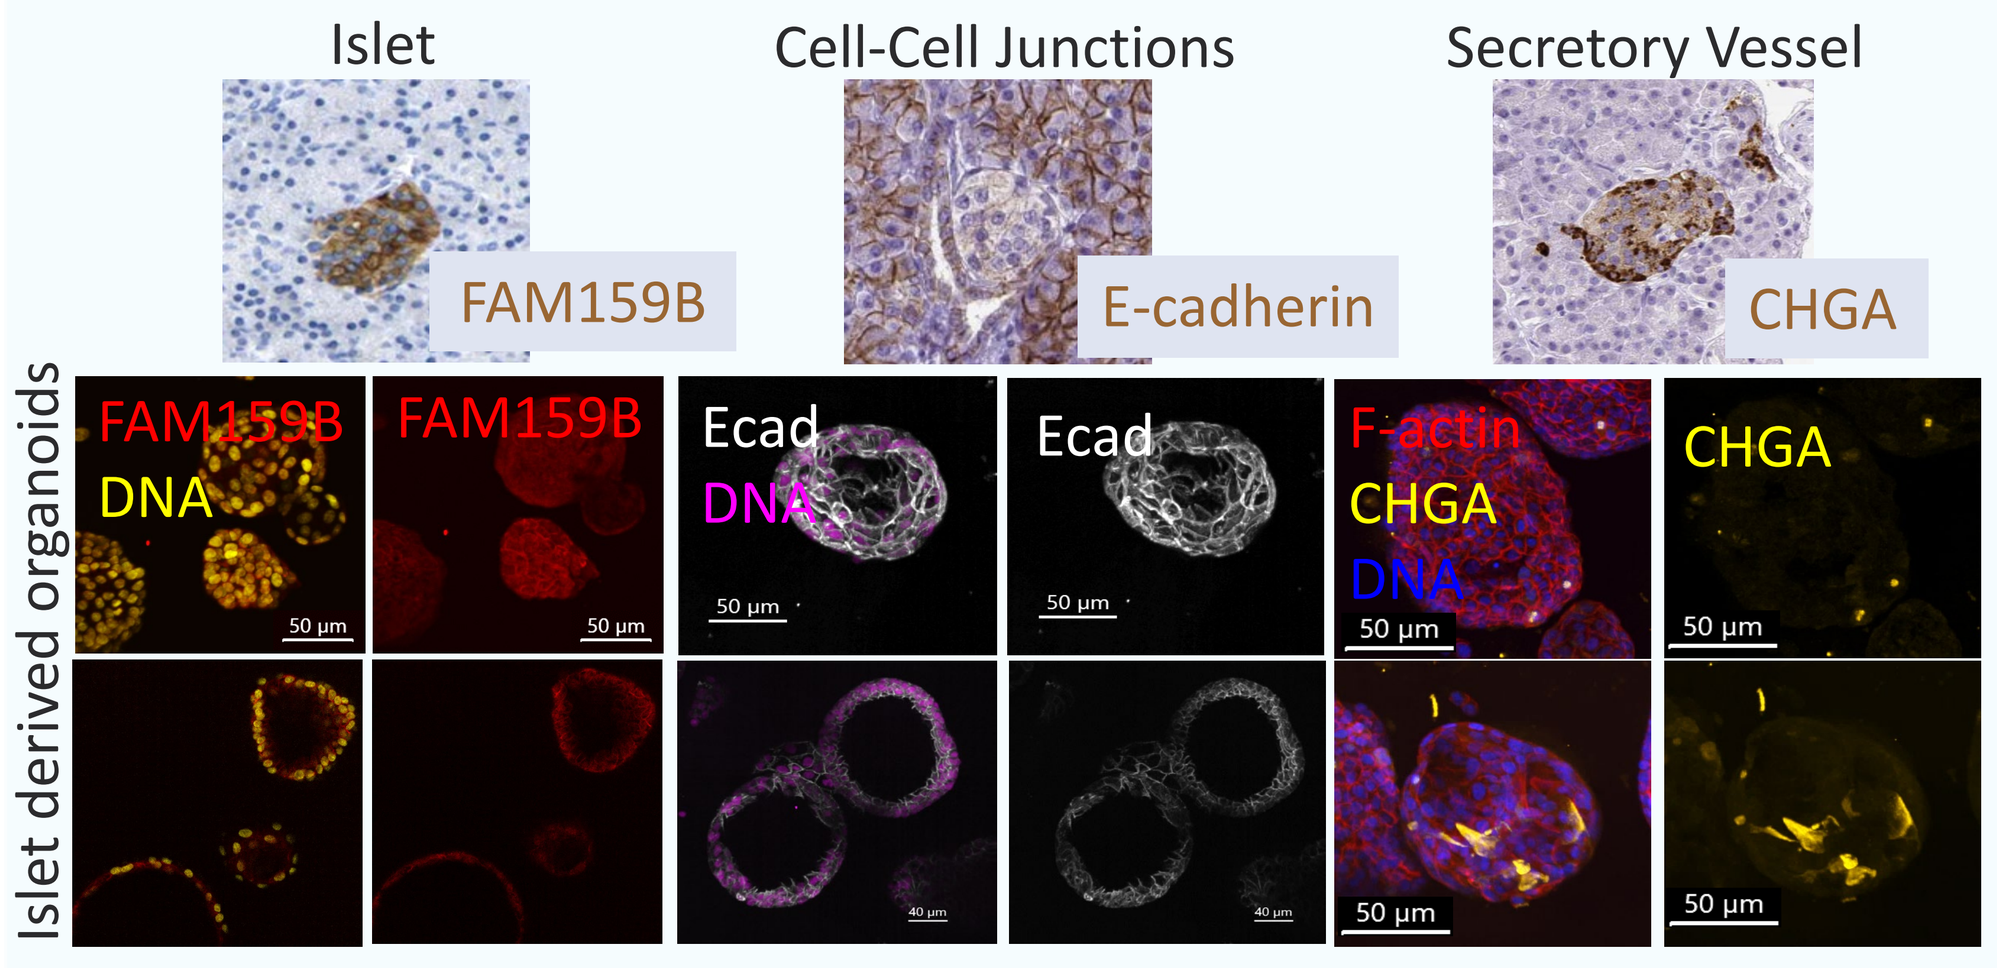
Islet
Cell-Cell Junctions
Secretory Vessel
FAM159B
E-cadherin
CHGA
Islet derived organoids
DNA
Ecad
F-actin

Figure 4: Pancreatic organoids were generated from freshly isolated human islets (A) and expression of islet markers was observed via immunocytochemistry (B). Islet marker FAM159B (red, left) and cell junction marker E-cadherin (white, middle) were expressed throughout pancreatic organoids. Chromogranin A (CHGA, yellow, left) was expressed in discrete regions, supporting a secretory phenotype.

Organoids Produce Functional β-Cell Markers: Pro-Insulin, C-Peptide, and Insulin

(A) Functional β–Cell Proteins Localize as Expected

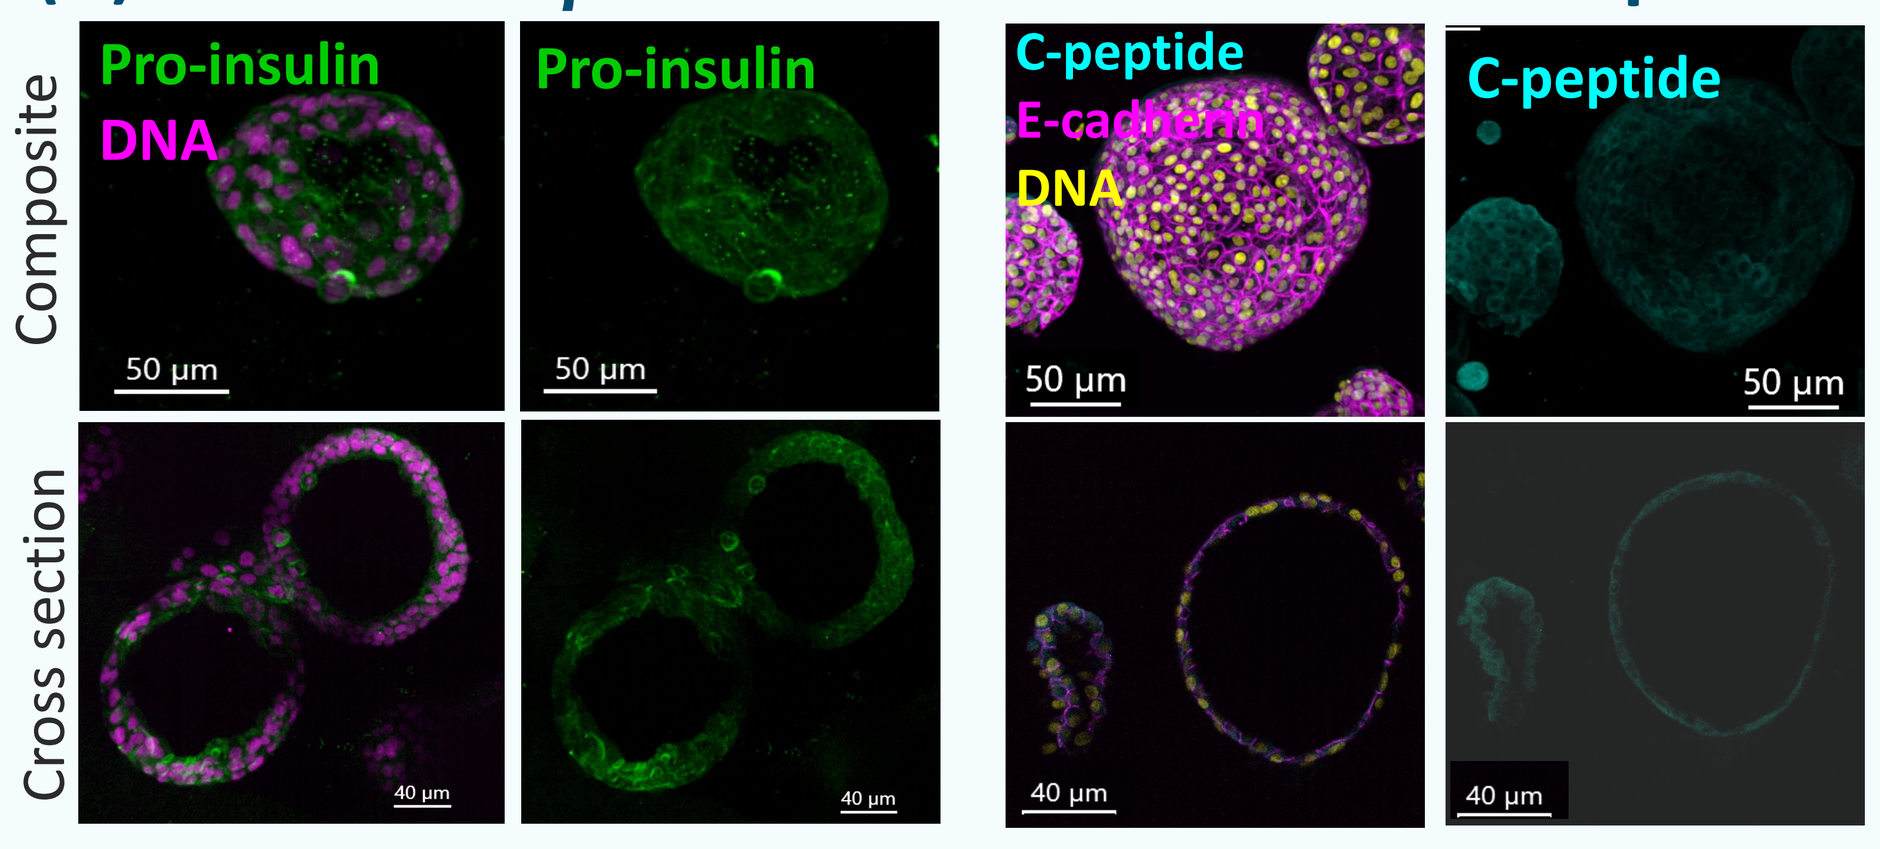
Composite
Cross section
Pro-insulin
DNA
C-peptide
E-cadherin

(B) Secreted Insulin is Maintained

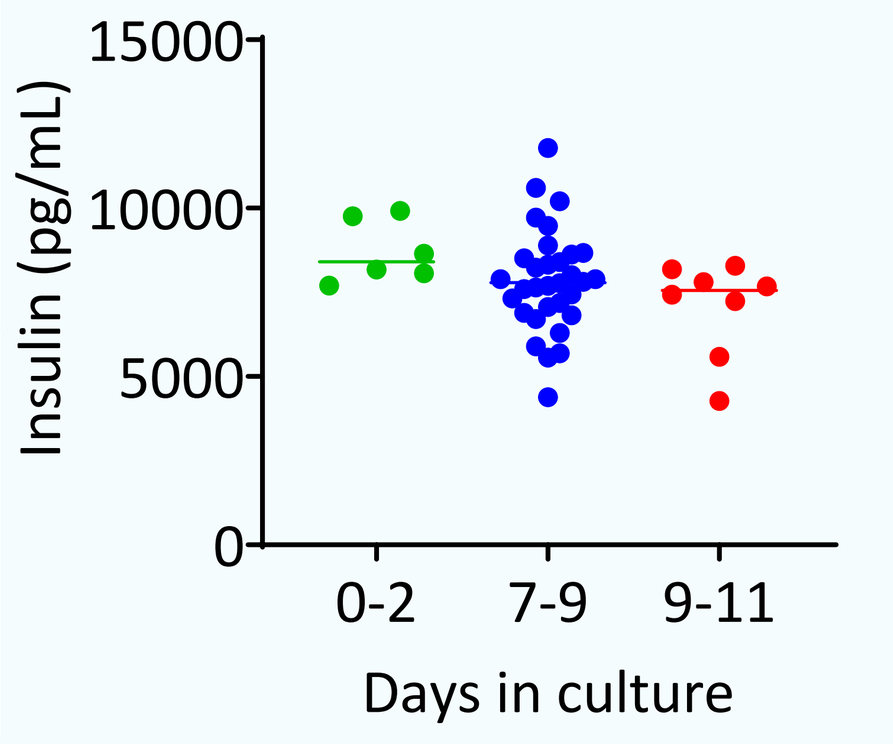
Insulin (pg/mL)
0-2
7-9
9-11
Days in culture

(C) Semi-Quantitative Assessment of β Cell Prevalence and Functional Proteins

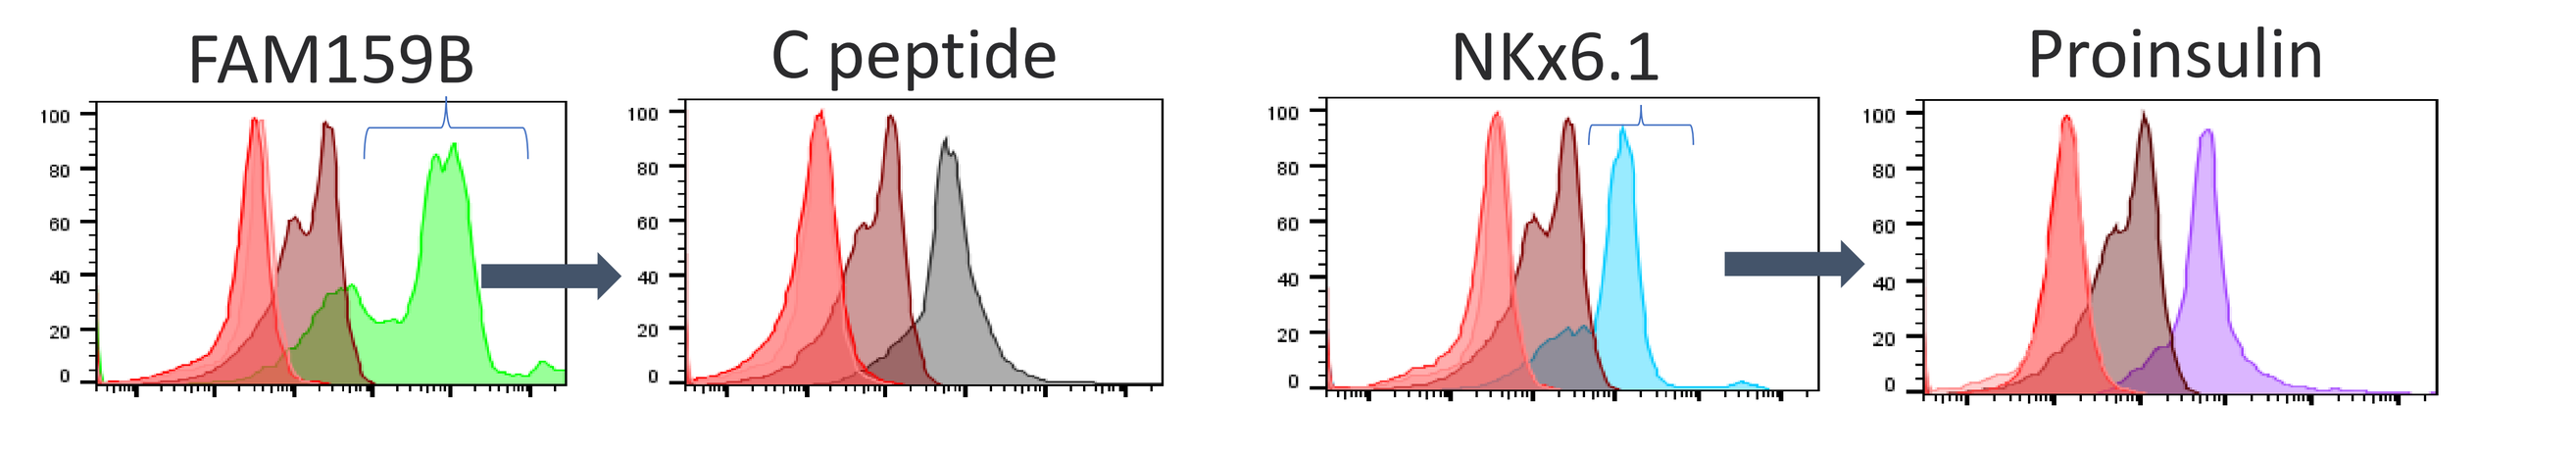
FAM159B
C peptide
NKx6.1
Proinsulin

Figure 5: Pancreatic organoids have islet specific functional markers. (A) Organoids expressed β-Cell-specific markers Proinsulin (green, upper left) and C-peptide (cyan, upper middle), as detected by immunocytochemistry. (B) β-Cell secreted insulin was detected in pancreatic organoid culture supernatants for the duration of the experiment by immunoassay. (C) FAM159B (green) or NKx6.1 (cyan) positive islet cells also expressed the β-Cell functional markers C-peptide (black) and Pro-insulin (purple) as determined by flow cytometry. FMO and negative controls are shown in red.

Autoantigenic Peptide Stimulated T1D Patient PBMC Increase β-Cell Death in Pancreatic Organoids

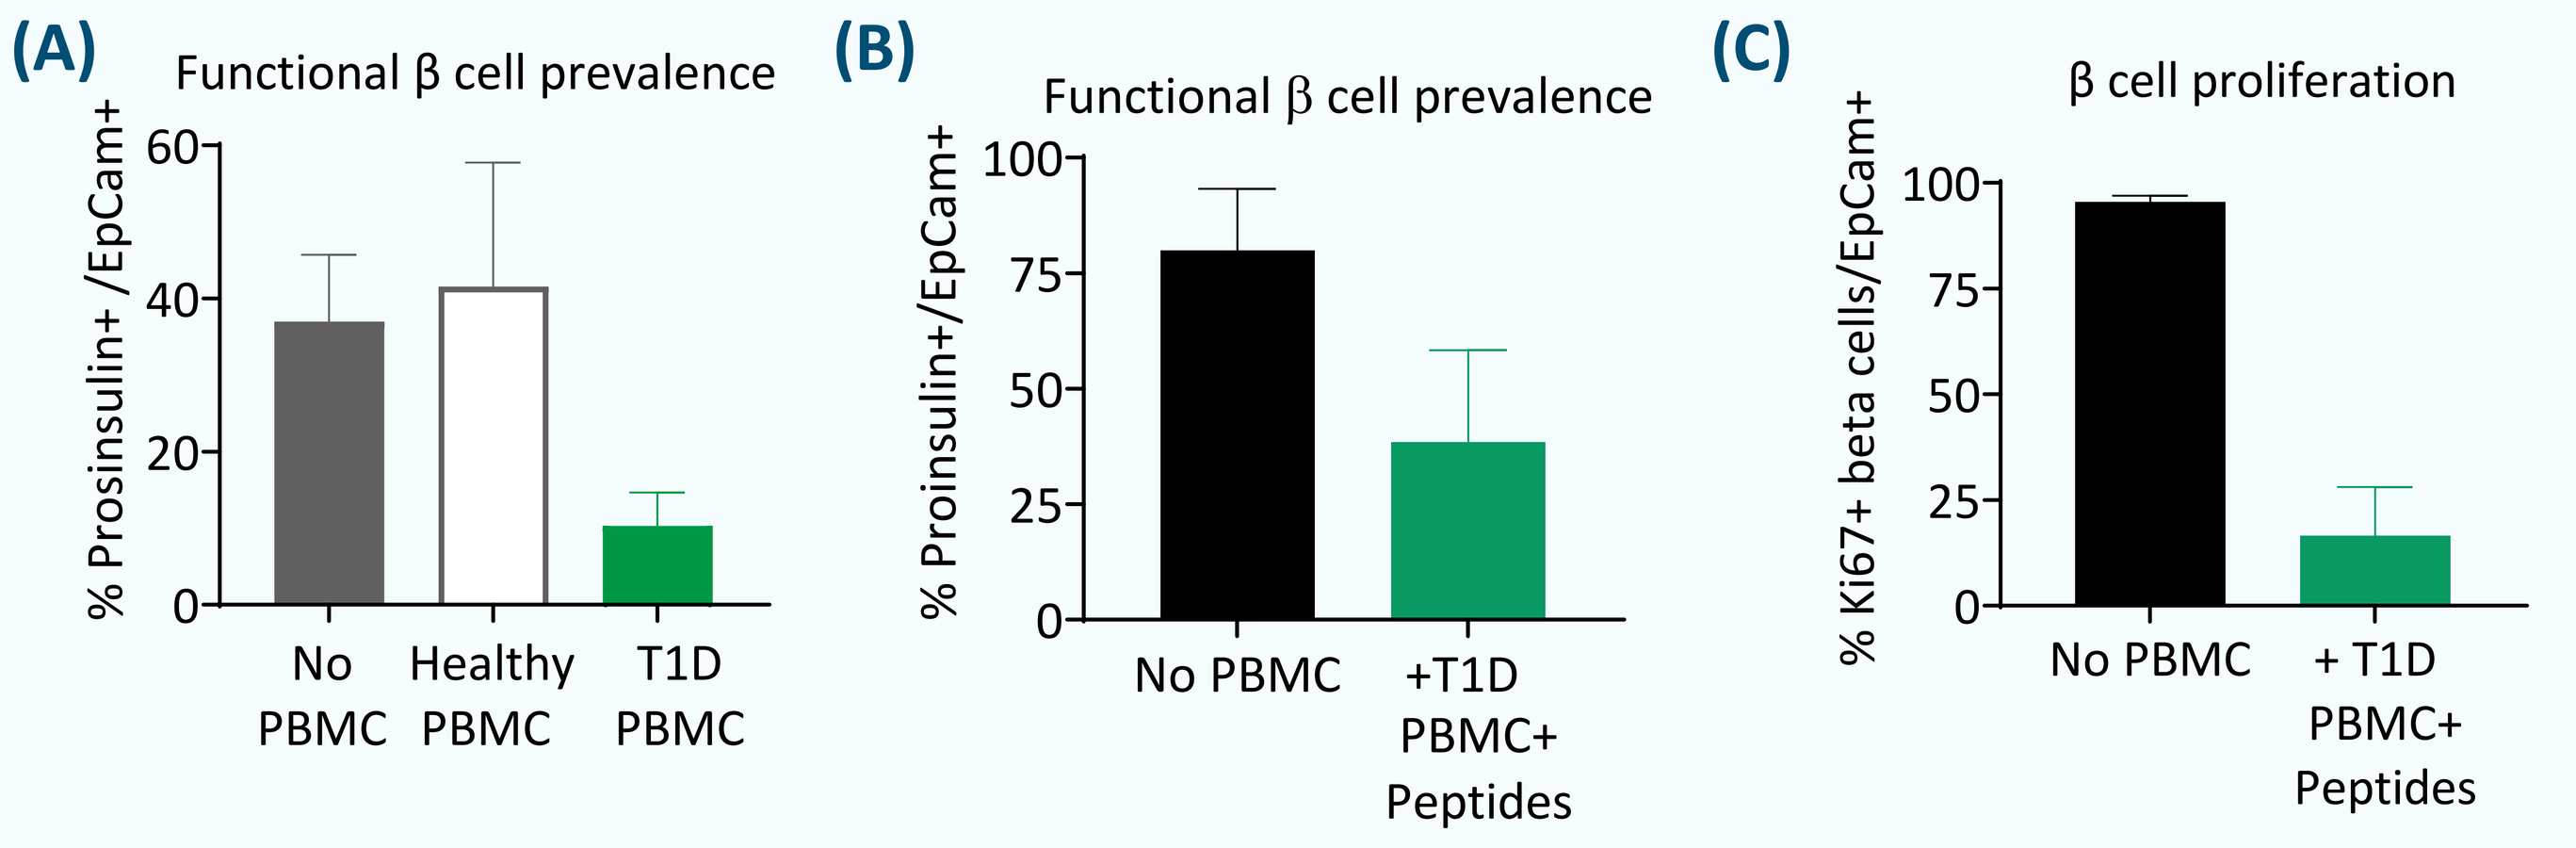
(A)
Functional β cell prevalence
% Prosinsulin+ /EpCam+
No PBMC
Healthy PBMC
T1D PBMC
(B)
Functional β cell prevalence
% Proinsulin+/EpCam+
No PBMC
+T1D PBMC+ Peptides
(C)
β cell proliferation
% Ki67+ beta cells/EpCam+
No PBMC
+ T1D PBMC+ Peptides

Figure 6: Assessment of pancreatic islet tissue destruction. (A) Pancreatic organoids co-cultured with PBMC from T1D patients had reduced functional β-Cells compared to healthy donor PBMC. (B) Autoantigenic peptides increased β-Cell death compared to organoids cultured in the absence of PBMC and (C) reduced β-Cell proliferation

Acknowledgements: The authors would like to thank the PBMC and pancreatic organ donors, and Dr. Calvin Kuo's laboratory (Stanford University; Palo Alto, CA) for assistance with the organoid methods.
Contact: Questions can be directed to Kristine Swiderek, Chief Development Officer, Mozart Therapeutics kswiderek@mozart-tx.com or by visiting the website at https://www.mozart-tx.com/
References: Li, et al. KIR + CD8 + T cells suppress pathogenic T cells and are active in autoimmune diseases and COVID-19. *Science,* 2022. DOI: 10.1126/science.abi9591

T1D PBMC + Autoantigenic Peptides Induce β-Cell Death and Diabetogenic Cytokines

(A) Diabetogenic Cytokines Induce β-Cell Death

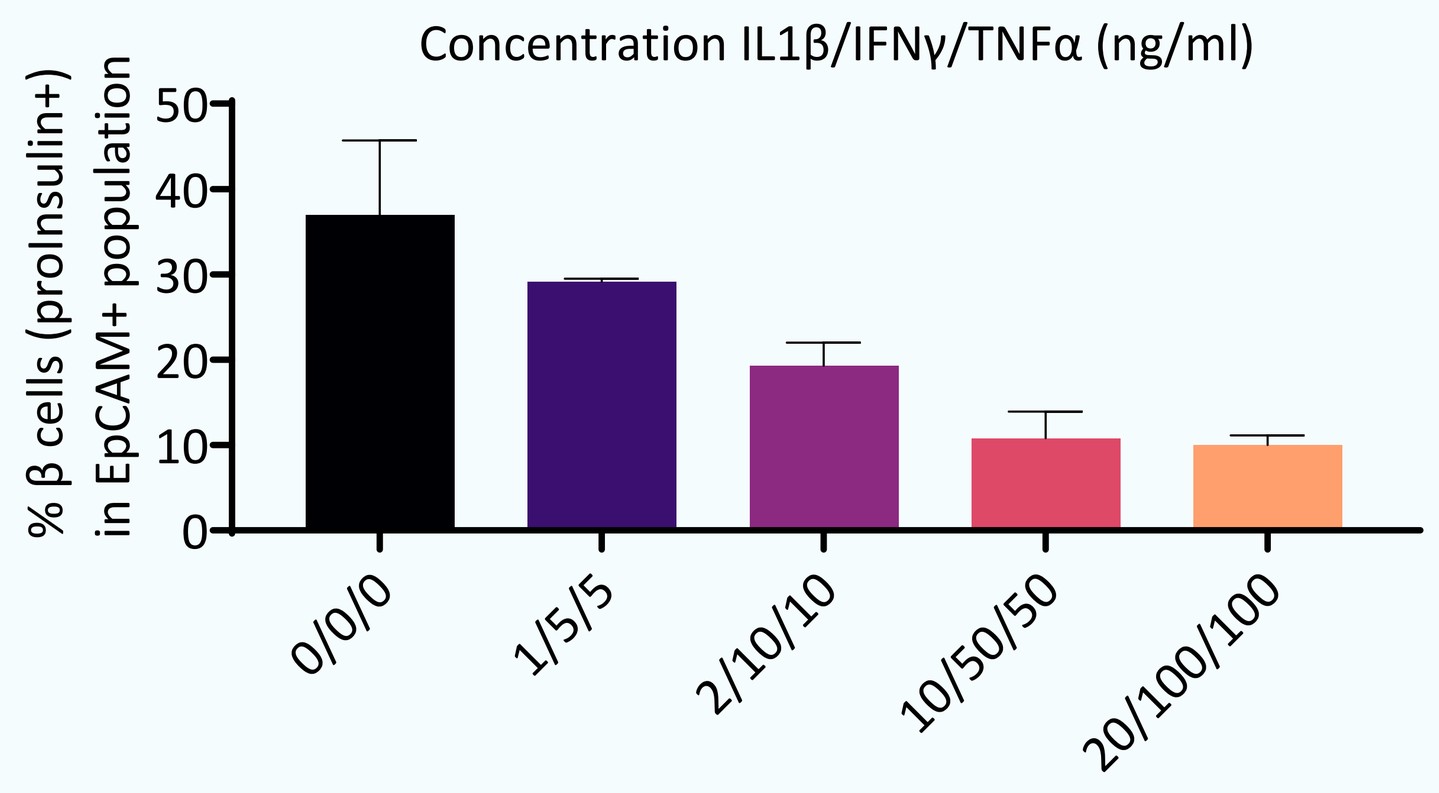
Concentration IL1β/IFNγ/TNFα (ng/ml)
% β cells (proInsulin+) in EpCAM+ population
0/0/0
1/5/5
2/10/10
10/50/50
20/100/100

(B) Peptide Pools Induce Diabetogenic Cytokines

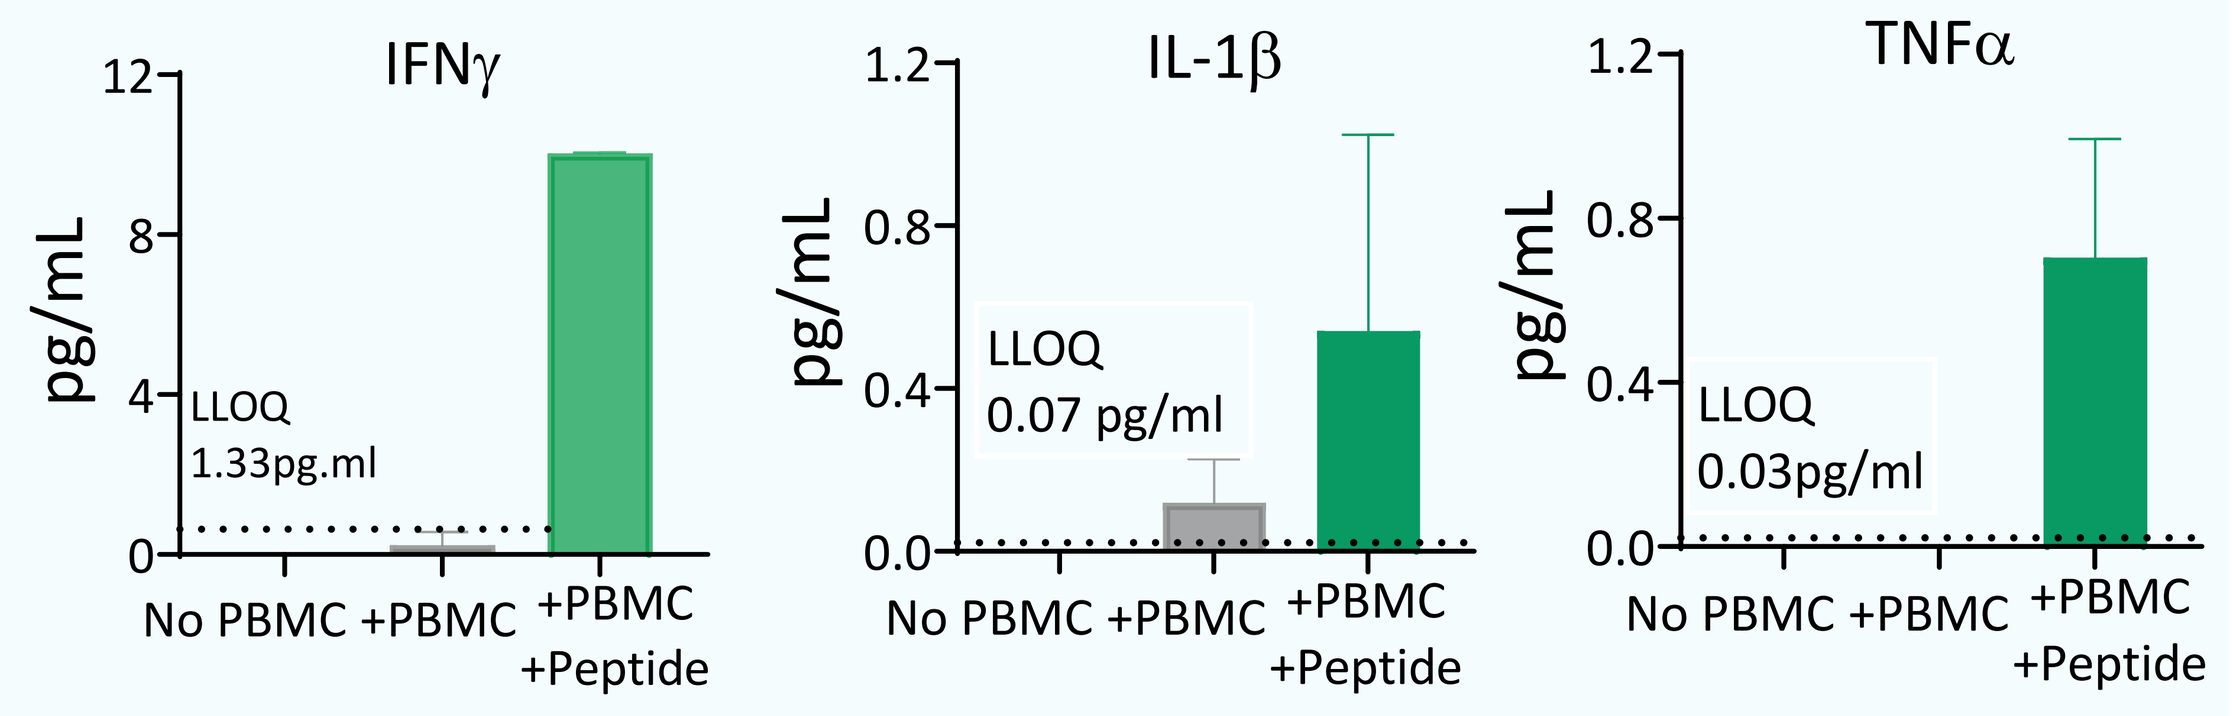
IFNγ
IL-1β
TNFα
pg/mL
LLOQ 1.33pg.ml
LLOQ 0.07 pg/ml
LLOQ 0.03pg/ml
No PBMC
+PBMC
+PBMC +Peptide

Figure 7: **Diabetogenic cytokines as a possible driver for β-Cell death.** (A) Pancreatic organoids cultured with increasing concentrations of diabetogenic proinflammatory cytokines caused a loss of viable Proinsulin+ β-Cells. (B) T1D PBMC+ organoid cocultures were stimulated with autoantigenic peptides, and increased secretion of IFNγ, IL1β, and TNFα, a possible driver for β-Cell death in Fig.6.

RESULTS AND CONCLUSIONS

- CD8 Treg from T1D patients appear reduced in prevalence and dysfunctional
- Ex vivo T1D organoid cultures allow complex assessment of in tissue immune responses
- CD8 Treg fail to control pathogenic T cell activation and cytokine production in T1D PBMC, and β–Cell damage in pancreatic organoids
- CD8 Treg functions in T1D PBMC can be restored
- Our data support the development of therapeutics designed to restore CD8 Treg functions in patients with T1D